Poster #3544

Phase I Study of Bortezomib and ^{153}Sm-lexidronam Combination for Refractory and Relapsed Multiple Myeloma

James R Berenson, MD[1,2], Regina A Swift[1], Russell Mapes[1] and Christina Abaya[2]

[1]Institute for Myeloma & Bone Cancer Research and [2]Oncotherapeutics, Inc., West Hollywood, CA, 90069

I. Abstract

Background: Multiple myeloma (MM) is a highly radiosensitive B-cell malignancy and radiation therapy is an effective treatment for these patients. Recent preclinical studies have demonstrated that the bone-seeking radionuclide, Samarium Sm153 lexidronam (Sam) in combination with the proteasome inhibitor, bortezomib (Vel), can synergistically inhibit proliferation of myeloma cell lines *in vitro* and reduce MM growth in mice bearing human MM without significant myelotoxicity. These results provide the basis for a new targeted therapeutic approach for refractory and relapsed MM patients involving combining Vel and Sam to improve the anti-MM effects of these agents without increasing their toxicity.
Aims: The primary objective of this dose escalation Phase I study is to determine safety and tolerability as well as the response rate as determined by Blade criteria of Vel + Sam treatment for patients with relapsed or refractory MM.
Methods: MM patients who had failed ≥2 prior treatments will be enrolled on this Phase I dose-escalation trial which involves six cohorts with three patients each. Previous treatment with Vel is allowed. Dose escalations in parallel arms are as follows:

Arm 1	Sam	Vel	Arm 2	Sam	Vel
Cohort 1	0.25 mCi/kg	1.0 mg/m^2	Cohort 4	0.25 mCi/kg	1.3 mg/m^2
Cohort 2	0.5 mCi/kg	1.0 mg/m^2	Cohort 5	0.5 mCi/kg	1.3 mg/m^2
Cohort 3	1.0 mCi/kg	1.0 mg/m^2	Cohort 6	1.0 mCi/kg	1.3 mg/m^2

A complete treatment cycle is 8 weeks and patients are to receive a maximum of 4 cycles. Vel is given on days 1, 4, 8 and 11 followed by a 45-day rest period. Sam is administered only on day 3. The cycle is repeated on Day 57 if disease is stable or improved and platelets and neutrophils recover to at least Grade 1 toxicity (may be delayed for up to four weeks). DLT is defined as cycle 1 grade 4 hematologic or Grade ≥3 non-hematologic toxicity.
Results: Eighteen patients have been enrolled to date in cohorts 1-5. Three patients have been treated in cohorts 1, 2, 4, and 5 and 6 patients have been entered in cohort 3. Fifteen patients are evaluable for response and the other 3 have not yet completed a treatment cycle. Of the 15 evaluable patients, 3 patients including one Vel refractory patient have shown responses [immunofixation+complete response {IF+CR} (n=2) and minor response (n=1)] three have stable disease and 9 patients showed progressive disease. One Vel refractory patient with progressive disease initially showed a transient IF+CR.

Results (cont'd): Two patients developed transient Grade 4 thrombocytopenia. One of these episodes occurred during Cycle 1 in a patient in cohort 3; and, as a result, cohort 3 has been expanded to a total of 6 patients. Four patients showed transient Grade 3 neutropenia. Only 2 patients showed treatment emergent neuropathy one of which resolved while on study. To date, seven patients remain on study and one patient has completed four cycles of therapy.

II. Materials and Methods

Patient Population:
Male or female patients who are 18 years of age or older with relapsed or refractory MM and have measurable disease.

Inclusion Criteria:
•Previously diagnosed with MM based on standard criteria (Durie, *Semin. Oncol.*, 1986; 13: 300-9)
•Currently has MM with either:
-Measurable disease, defined as: monoclonal immunoglobulin spike on serum electrophoresis of ≥1 gm/dL and/or urine monoclonal immunoglobulin spike of ≥200 mg/24 hrs or evidence of lytic bone disease.
-Non-measurable disease (i.e. patients with nonsecretory or oligosecretory MM).
•Relapsed following a response or stable disease to chemotherapy or high-dose chemotherapy, or refractory to their most recent chemotherapy, whether or not containing systemic corticosteroids
•Baseline platelets ≥75 x 10^9/L and ANC ≥1.5 x 10^9/L

Exclusion Criteria:
•POEMS syndrome and skin changes
•Active infection
•Serum calcium ≥ 14 mg/dL
•Major surgery within four weeks
•Chemotherapy within three weeks
•Corticosteroids (>10 mg/day prednisone or equivalent) within three weeks
•Immunotherapy, antibody or radiation therapy within 4 weeks
•Known HIV history or active hepatitis B or C viral infection
•Baseline >grade 1 neuropathy
•Extramedullary myeloma
•History of allergic reaction attributable to compounds of similar chemical or biological composition to bortezomib, boron, mannitol, EDTMP or phosphonates

Study Treatment Schedule

- Each treatment cycle is 8 weeks
- Maximum of 4 cycles permitted

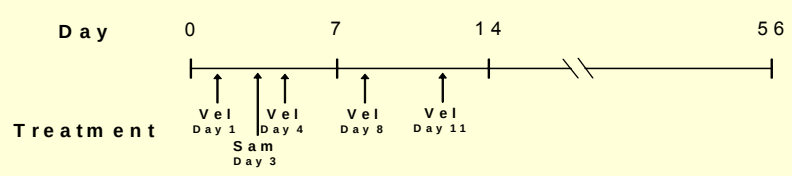

Day
0
7
14
56
Treatment
Vel
Day 1
Vel
Day 4
Vel
Day 8
Vel
Day 11
Sam
Day 3

III. Results (updated as of 12/6/06)

Demographics and Treatment History

Patient #	Cohort	Age	Gender	# Prior Myeloma Tx's	Prior Bortezomib
1	1	68	M	5	Y
2	4	61	M	5	Y
3	1	48	M	3	Y
4	4	68	F	4	Y
5	1	55	F	2	N
6	4	66	M	1	N
7	2	69	F	8	Y
8	5	63	M	6	Y
9	2	66	F	1	N
10	5	62	F	2	N
11	2	51	M	2	Y
12	5	62	M	3	Y
13	3	73	M	3	Y
14	3	69	M	7	Y
15	3	78	F	7	Y
16	3	71	M	2	N
17	3	46	M	2	N
18	3	66	M	2	N
19	6	68	M	4	N
20	6	63	M	1	Y

Response to Study Treatment

Cohort 1 (0.25 – 1.0)[1]		# Tx Cycles	Cohort 4 (0.25 – 1.3)[1]		# Tx Cycles
Pt. 1	PD	1	Pt. 2	PD	1
Pt. 3	PD	1	Pt. 4	SD	2
Pt. 5	PD	1	Pt. 6	CR*	4
Cohort 2 (0.5 – 1.0)[1]		**# Tx Cycles**	**Cohort 5 (0.5 – 1.3)[1]**		**# Tx Cycles**
Pt. 7	PD	1	Pt. 8	SD	4
Pt. 9	CR*	2	Pt. 10	SD	3
Pt. 11	PD	1	Pt. 12	MR	4
Cohort 3 (1.0 – 1.0)[1]		**# Tx Cycles**	**Cohort 6 (1.0 – 1.3)[1]**		**# Tx Cycles**
Pt. 13	PD	1	Pt. 19	NYE	1
Pt. 14	PD	1	Pt. 20	NYE	1
Pt. 15	PD	1			
Pt. 16	PD	1			
Pt. 17	PD	1			
Pt. 18	SD	2+			

* Not confirmed by bone marrow exam per patient choice

NYE – Patient currently on study not yet evaluable for response

[1](Sm-153, mCi/kg - Bortezomib, mg/m^2)

+ indicates patient still on study

Grade 3 and 4 Adverse Events (20 patients, 35 treatment cycles)

Adverse Event	Grade 3	Grade 4
Altered Mental Status	1*	
Anemia	1	
Bone Pain	1	
Dyspnea	1*	
Headache	2*	
Leg Cramps	1*	
Muscle Weakness	1*	
Neutropenia	4	1
Renal Failure	1*	
Thrombocytopenia	0	2
Thrombosis	1*	
Neuropathy[a]	0	0

[a]Two patients with Gr. 1 neuropathy at baseline progressed to Gr. 2, one case resolved while on Tx

*Judged to be result of progressive disease and not related to study medications

IV. Conclusions

- The combination of Samarium-153 lexidronam and bortezomib appears to be well-tolerated at the doses studied.
- The primary treatment-related toxicity has been a transient decrease in platelets and neutrophils.
- The incidence of treatment emergent neuropathy has been low and the severity mild to moderate.
- One instance of DLT was observed in cohort 3.
 - Expansion of the cohort resulted in no additional DLTs
- Among the 18 patients evaluable for response:
 - 3 have responded (2 CR and 1 MR)
 - 4 have shown stable disease.
- Patients enrolled to the last cohort of this Phase I study continue to be evaluated, preparations are underway for a follow-on Phase II study.

Disclosure Statement

Employment: None Honoraria: None
Consultancy: None Paid Expert: None
Ownership: None Membership: None
Financial Relationship: None
Ownership Interest in a Publicly Traded Company: None
Research Funding: James R. Berenson, MD - research grants